Hyaluronic acid in the treatment of osteoarthritis of the shoulder

R.J Petrella

Canadian Centre for Activity and Aging, the University of Western Ontario, London, Ontario, Canada

Abstract N° FRI0343

Introduction

Osteoarthritis (OA) of the shoulder is a complex but nonetheless debilitating condition for many active older adults. In a previous pilot study, hyaluronic acid (HA) was shown effective in improving outcomes in patients with OA of the shoulder. This larger and longer term follow up describes efficacy and safety of HA in OA of the shoulder.

Objectives

To determine the effectiveness of intra-articular injection of HA on pain and function in patients with unilateral osteoarthritis of the acromio-clavicular (AC) shoulder joint.

Methods

682 patients with primary unilateral osteoarthritis of the AC joint received intra-articular HA in the shoulder (AC) between 1999 and 2007. They were administered 3-weekly intra-articular injections of hyaluronic acid (Suplasyn 2.0 ml; 10 mg/ml). No other treatment modalities were administered during the two weeks prior to and during the study. Baseline assessment of pain and function included VAS with ROM and impingement testing and WOMAC-VAS for pain, stiffness and disability. Pain and function outcomes as well as patient global satisfaction (5-point categorical scale) and adverse events (AEs) were also collected at 4 weeks post-treatment and then similarly with each successive treatment series. Differences were compared using ANOVA and considered significant at $p<0.05$.

Results

No serious AEs were reported while 14 minor AEs included pain at the injection site, erythema of the injection site and nausea. Subjects were 61% male aged 57±10 years who received 6 consecutive series of treatment (range 6-9), every 37±7 weeks and used 2±1 concomitant oral or topical treatments following the first series.
Significant improvement in all indices of pain and function were observed using the WOMAC and VAS response to clinical testing. Resting VAS pain improved similarly between the first and final series (7.4±2 to 2.1±2, $p<0.05$, compared to 7.1±1.6 to 1.9±1.5, $p<0.05$ respectively). Injections were highly satisfactory (4±1) with no difference between the first and last series.

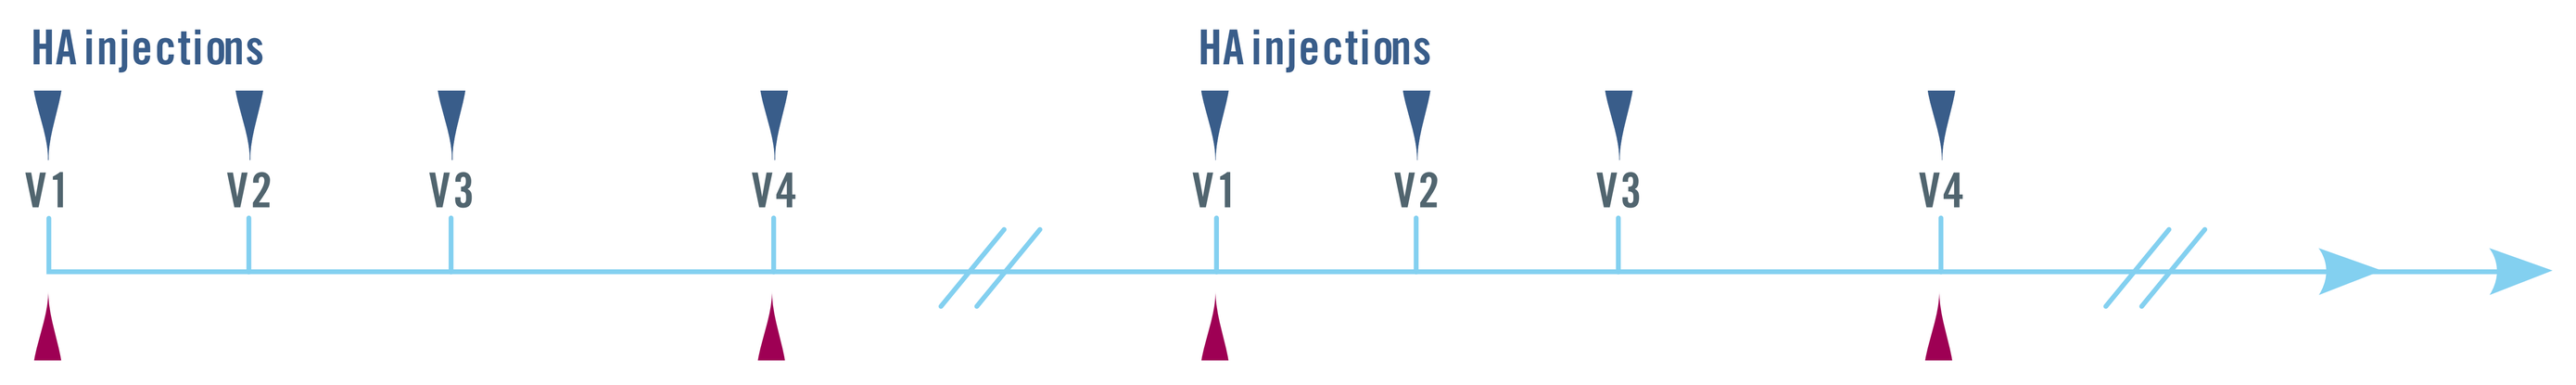

HA injections
V1
V2
V3
V4
HA injections
V1
V2
V3
V4

Evaluations V4 = 4 weeks post treatment

Baseline characteristics

Patients (n)	682
Age	57 ±10
Concomitant oral or tropical treatment (after 1st series)	2 ± 1
Number of series of intra-articular HA	6-9
Mean interval between series (weeks)	37 ± 7

Efficacy

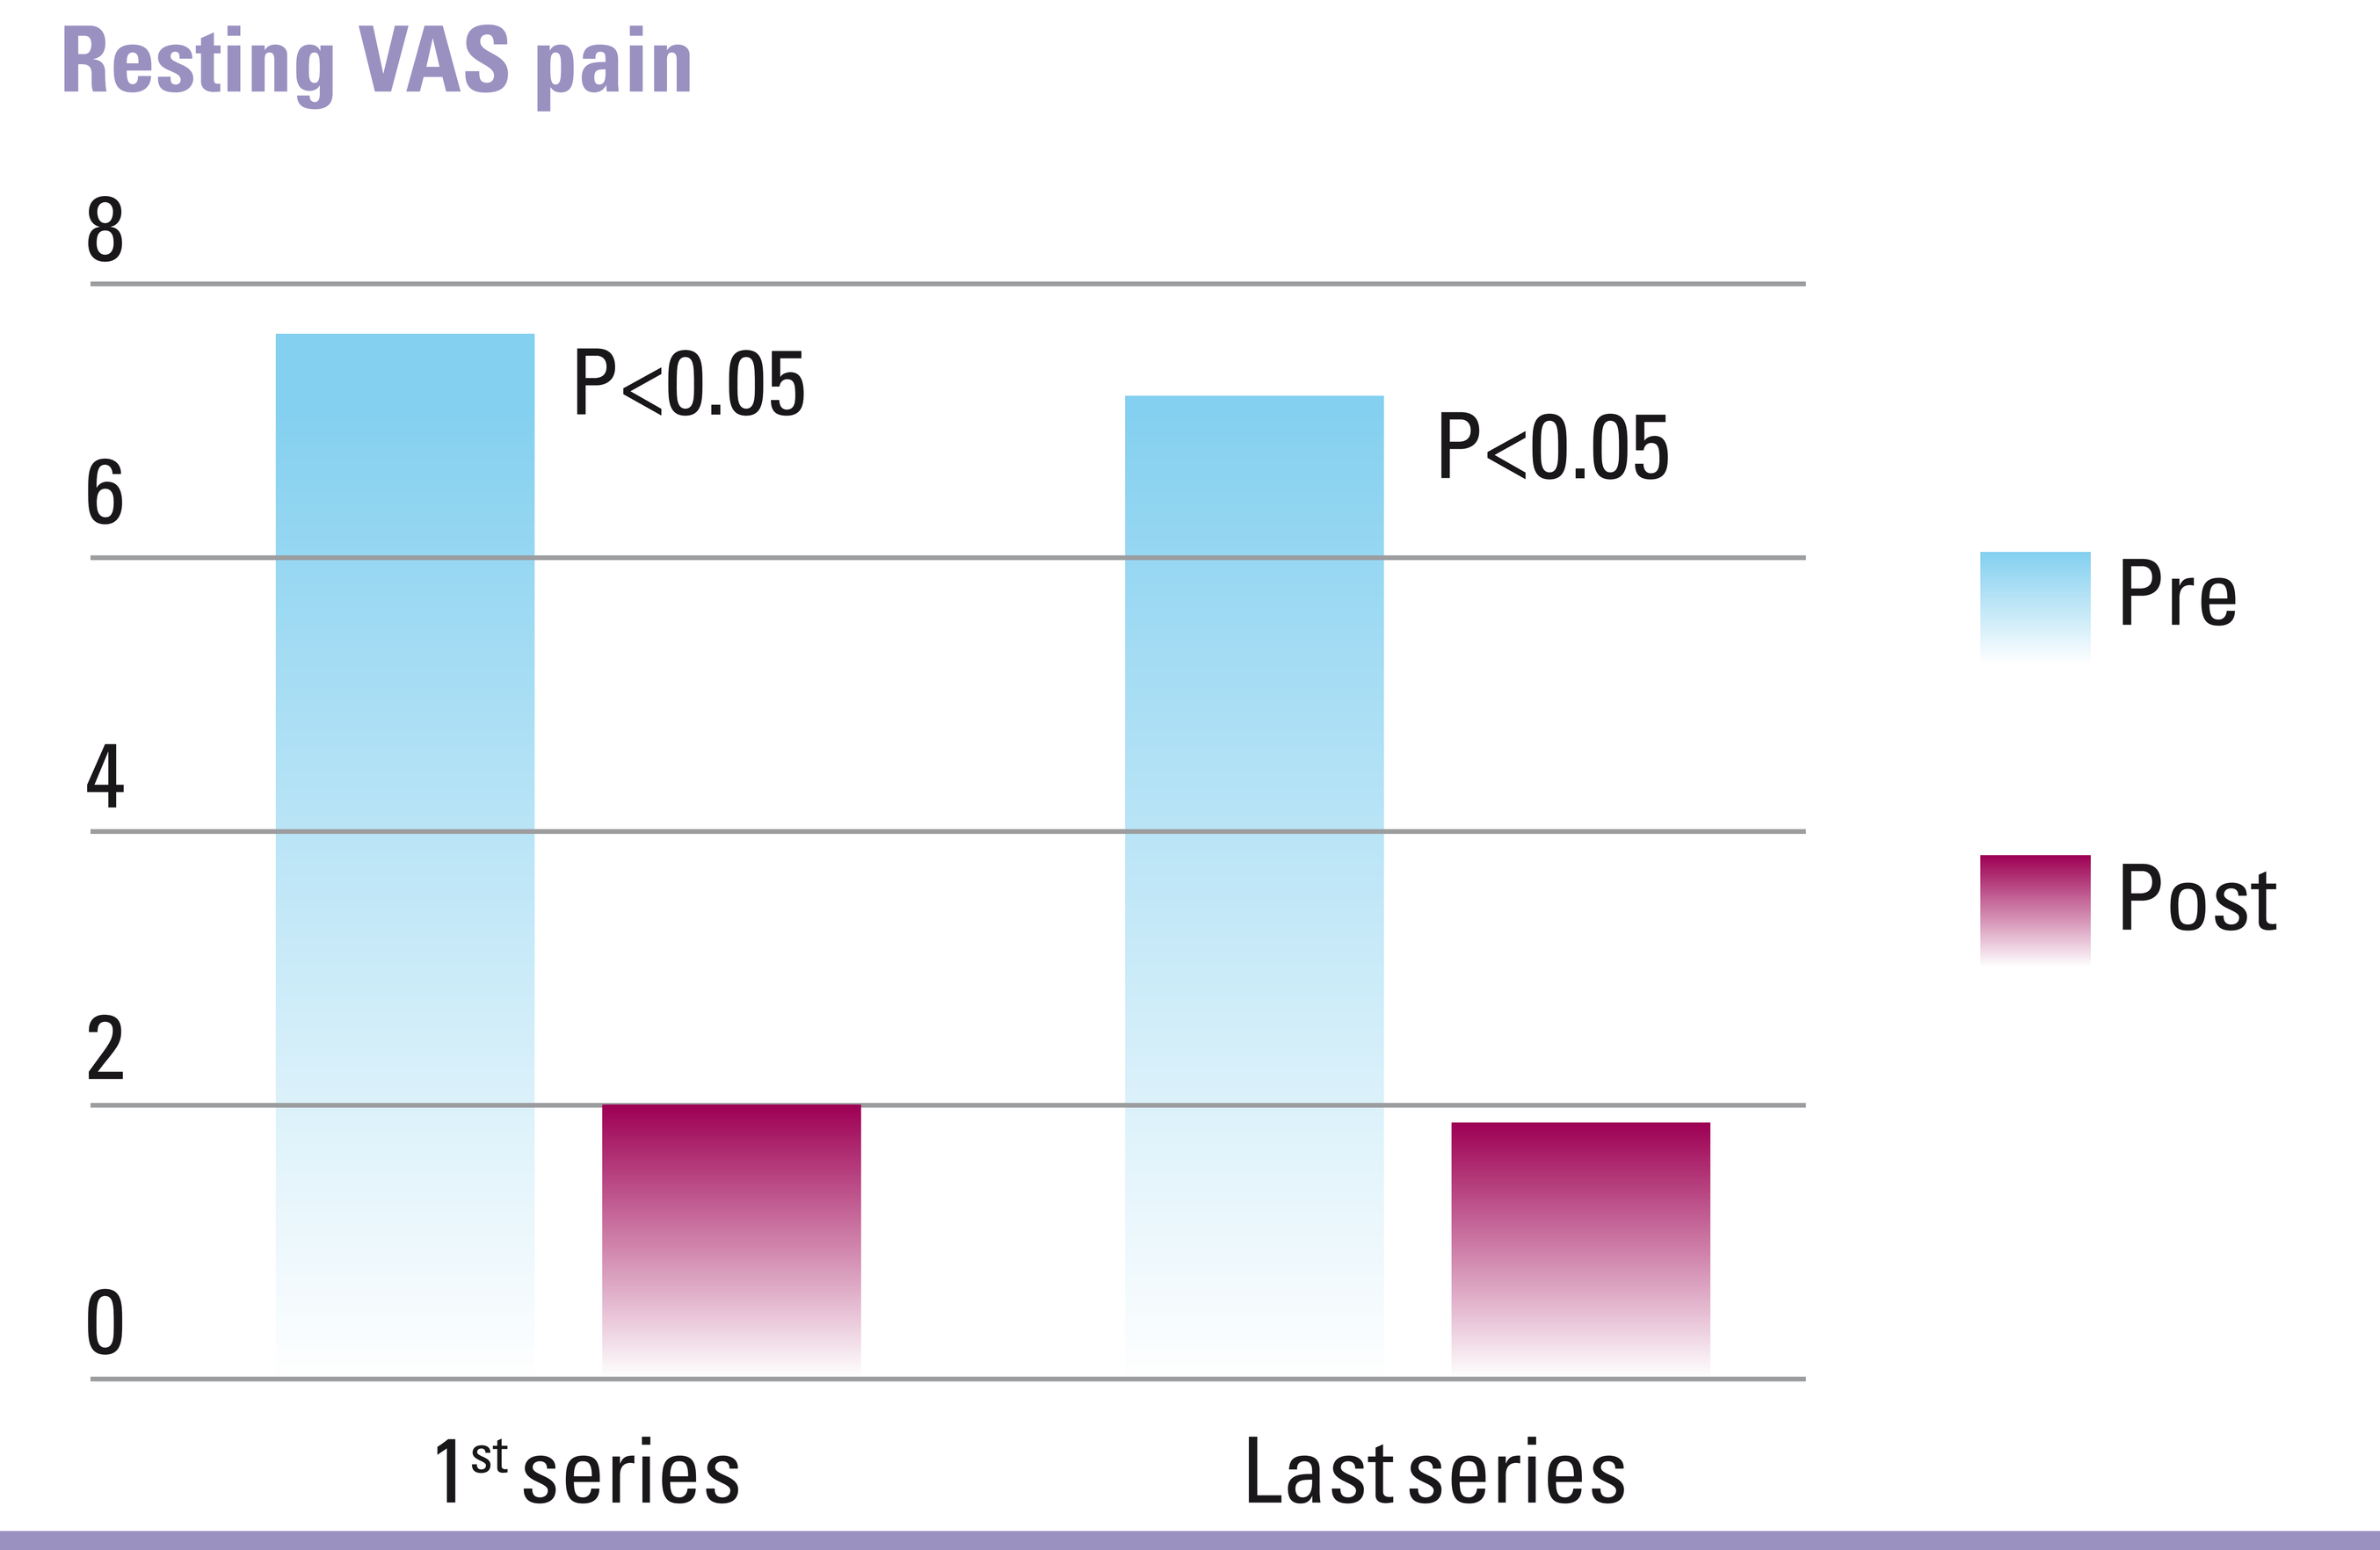

Resting VAS pain
8
6
4
2
0
P<0.05
P<0.05
Pre
Post
1st series
Last series

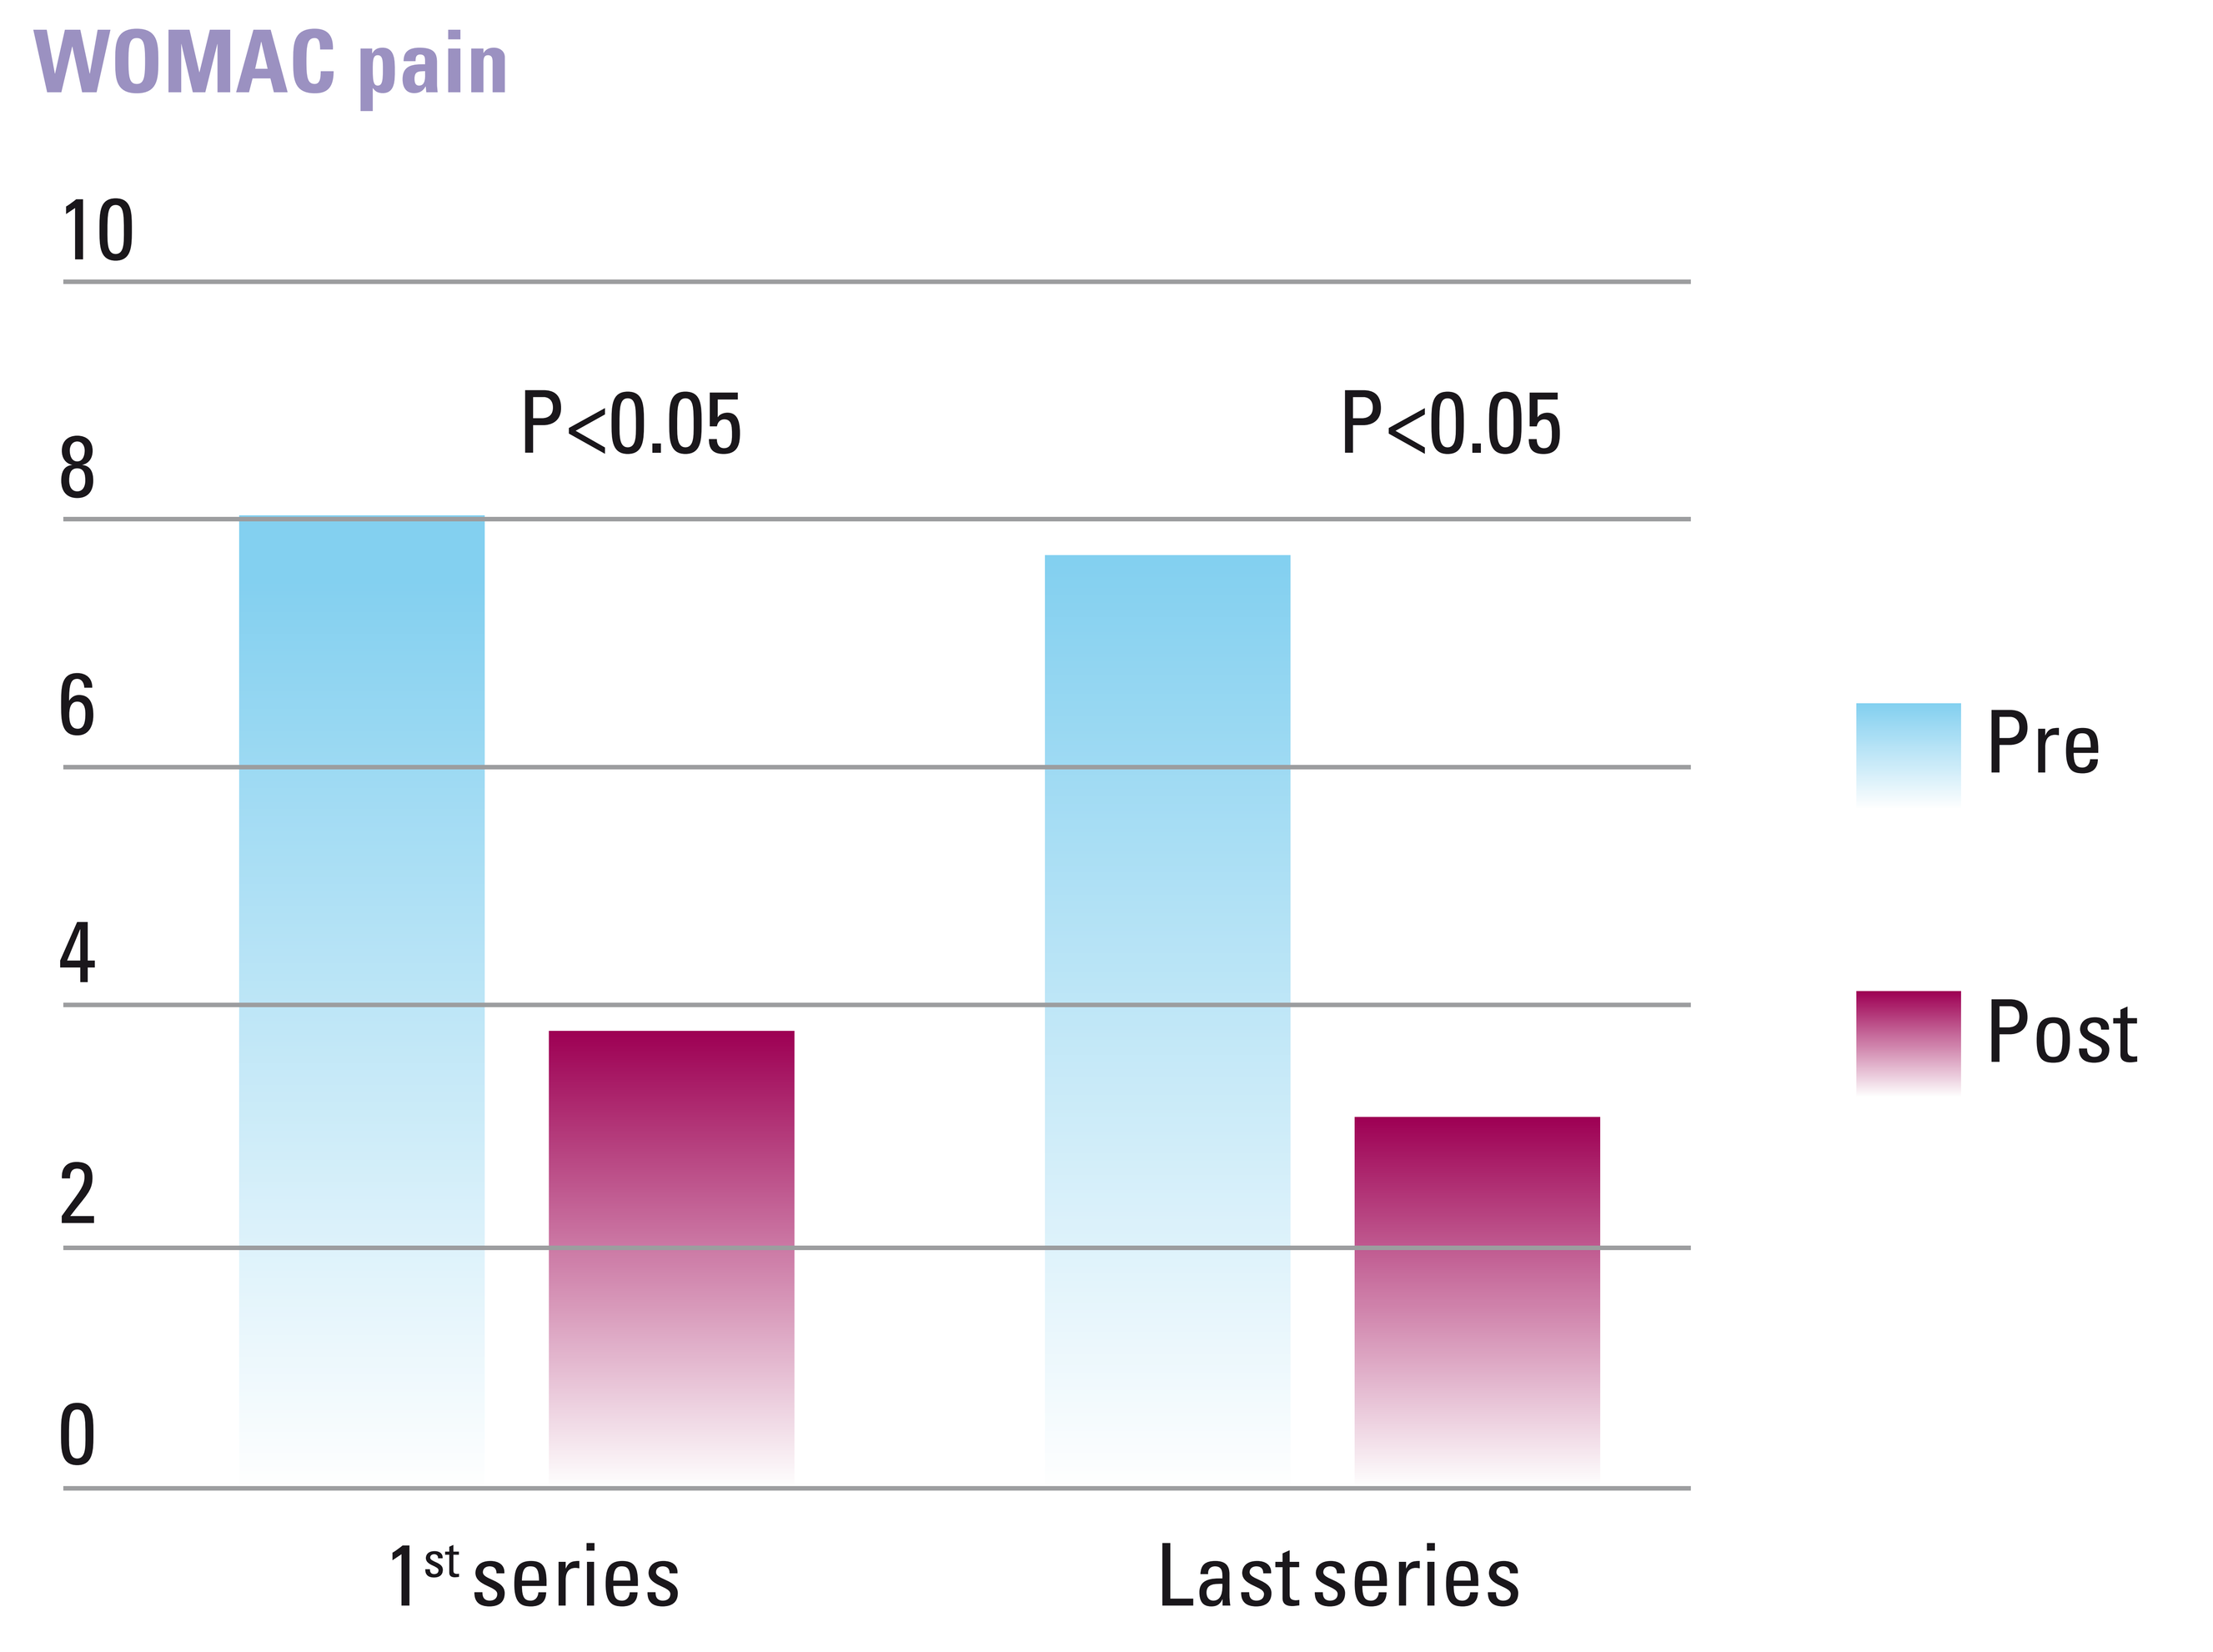

WOMAC pain
10
8
6
4
2
0
P<0.05
P<0.05
Pre
Post
1st series
Last series

Conclusions

➔ **Suplasyn injections resulted in a significant improvement in pain, stiffness and disability over at least 6 consecutive series, with very few AEs and was highly satisfactory to patients.**

➔ **Further assessment of efficacy compared to other modalities is needed.**

Safety

➔ No serious AE was reported
➔ 14 minor AEs including pain and /or erythema at injection site, nausea.

Patient satisfaction
➔ Injections were highly satisfactory, with no difference between the first and last series (4 ± 1).